5th International Congress on Drug Therapy in HIV Infection, Glasgow, UK (October 22-26, 2000)

Kaletra (ABT-378/ritonavir) in Antiretroviral-Naïve HIV+ Patients: Follow-up Beyond Two Years and Viral Load Suppression Below 3 Copies/mL

R. Stryker[1], S. Brun[12], M. King[12], T. Marsh[12], R. Murphy[2], C. Hicks[3], J. Eron[4], J. Thommes[1], R. Guilick[5], M. Glesby[5],
M. Thompson[6], C. White[7], C. Benson[8], M. Albrecht[9], H. Kessler[10], K. Real[12], A. Japour[12], L. Perrin[11], and E. Sun[12], for the M97-720 Study Group
[1]Pacific Oaks Research, [2]Northwestern, [3]Duke, [4]U. N. Carolina, [5]Cornell,
[6]AIDS Research Consortium of Atlanta, [7]Baylor/Thomas Street Clinic, [8]U. Colorado, [9]Harvard, [10]Rush, [11]U. Geneva, and [12]Abbott Laboratories

BACKGROUND

ABT-378 (lopinavir) is a novel HIV protease inhibitor (PI) that is co-formulated with ritonavir, an inhibitor of cytochrome P450 3A. It is exquisitely sensitive to pharmacokinetic enhancement by ritonavir, resulting in substantially increased ABT-378 drug exposure, even at low ritonavir doses. At the dosage selected for phase III clinical trials, 400 mg ABT-378/100 mg ritonavir BID (as 3 co-formulated capsules BID), ritonavir concentrations are below those required for antiviral activity.[1] The mean ABT-378 C_{trough}/EC_{50} ratio (Inhibitory Quotient or IQ) for wild-type HIV is ≥75 when dosed at 400/100 mg BID, potentially providing a pharmacologic barrier to the emergence of viral resistance and activity against resistant virus.[1] In contrast, the IQs for currently available PIs whether dosed as single PIs or in conjunction with ritonavir, range from 1-26 (Figure 1), based on the EC_{50} values determined in the presence of 50% human serum.[2] C_{trough} values in HIV+ patients for currently available PIs are derived from published sources including package inserts.[3-14]

The efficacy and safety of Kaletra (lopinavir/ritonavir, formerly known as ABT-378/r) are currently being studied in HIV-infected patients, both antiretroviral-naïve and PI-experienced. The M97-720 study is an ongoing phase II double-blind trial of ABT-378/r in combination with d4T and 3TC in antiretroviral-naïve patients. Results through Week 108 as well as results from an ultra-sensitive HIV RNA assay with a level of quantitation of ≤3 copies/mL are presented here.

Figure 1. C_{trough}/EC_{50} Ratio (Inhibitory Quotient) for PIs Given Alone or with RTV*

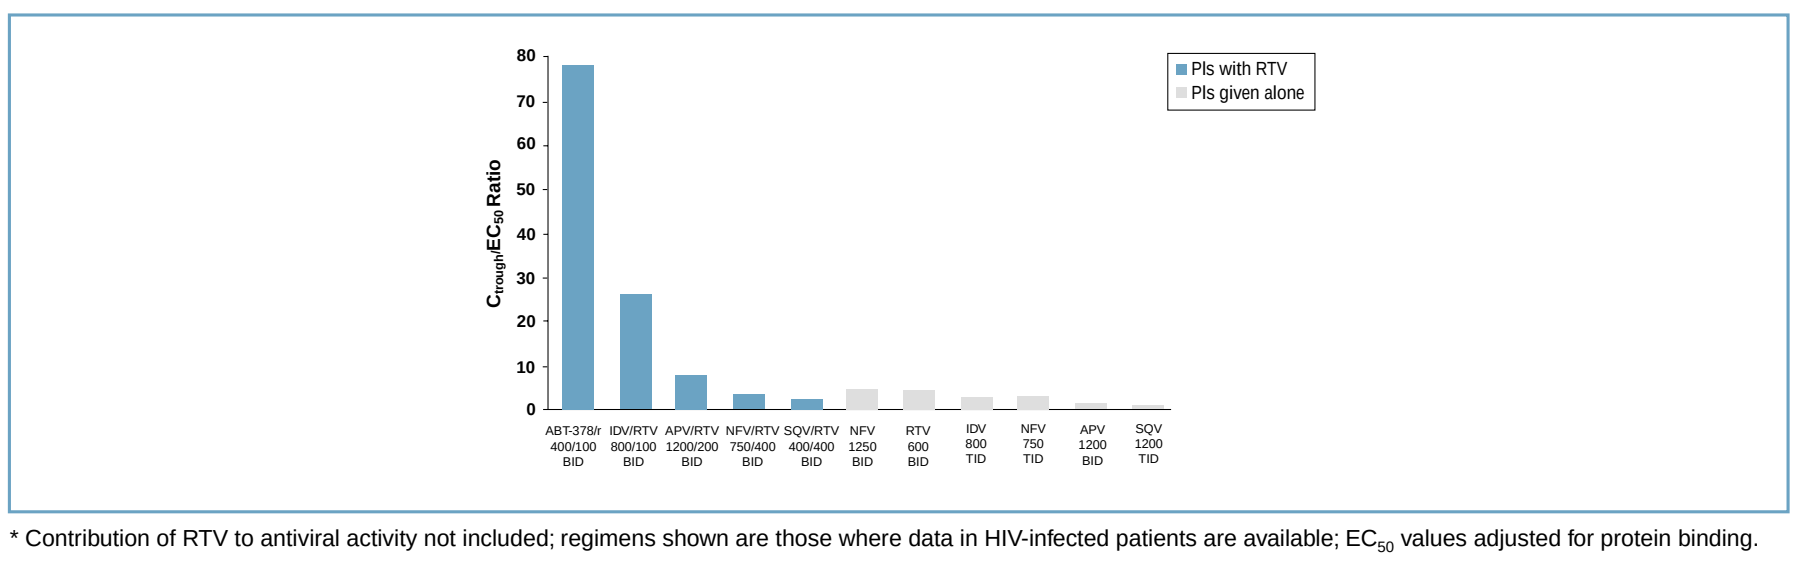
Ctrough/EC50 Ratio
80
70
60
50
40
30
20
10
0
PIs with RTV
PIs given alone
ABT-378/r 400/100 BID
IDV/RTV 800/100 BID
APV/RTV 1200/200 BID
NFV/RTV 750/400 BID
SQV/RTV 400/400 BID
NFV 1250 BID
RTV 600 BID
IDV 800 TID
NFV 750 TID
APV 1200 BID
SQV 1200 TID

* Contribution of RTV to antiviral activity not included; regimens shown are those where data in HIV-infected patients are available; EC_{50} values adjusted for protein binding.

METHODS

Entry Criteria

- Antiretroviral-naïve patients.
- Plasma HIV RNA ≥5,000 copies/mL with no CD4 cell count restriction.

Study Design and Analysis

- One hundred antiretroviral-naïve patients were randomized to receive one of three dosage levels of ABT-378/r (200/100 mg BID, 400/100 mg BID or 400/200 mg BID), together with d4T (40 mg BID) and 3TC (150 mg BID) given either after 3 weeks of monotherapy (Group I) or from study entry (Group II) (Figure 2).
- Enrollment into Group II began following an evaluation of preliminary efficacy and safety of ABT-378/r in Group I.
- After 48 weeks, all patients began conversion to open-label ABT-378/r 400/100 mg BID dosing.
- Plasma HIV RNA was quantified using Roche Amplicor HIV-1 Monitor™ (lower limit of quantitation [LLQ] 400 copies/mL) and the Roche Amplicor HIV-1 Monitor Ultrasensitive Quantitative PCR, Version 1.0 (LLQ 50 copies/mL). Samples from patients with HIV RNA <50 copies/mL at Weeks 24, 48, or 72 were analyzed using an experimental modification of the standard Roche Amplicor HIV RNA previously described by Perrin et al. This modified assay allows for a limit of quantitation of ≤ 3 copies/mL.[15]

Figure 2. M97-720 Study Schema

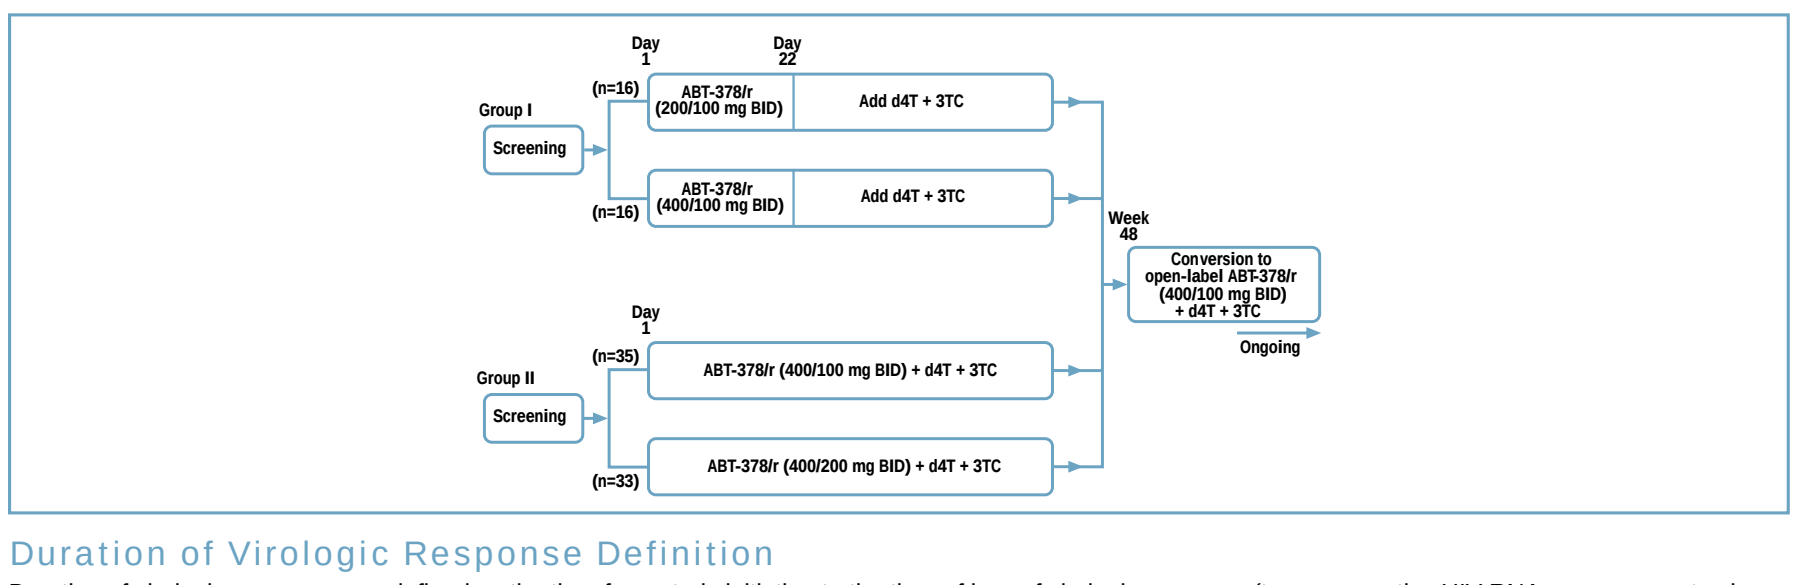
Day 1
Day 22
Group I
Screening
(n=16)
ABT-378/r (200/100 mg BID)
Add d4T + 3TC
ABT-378/r (400/100 mg BID)
Add d4T + 3TC
(n=16)
Week 48
Conversion to open-label ABT-378/r (400/100 mg BID) + d4T + 3TC
Ongoing
Day 1
Group II
Screening
(n=35)
ABT-378/r (400/100 mg BID) + d4T + 3TC
ABT-378/r (400/200 mg BID) + d4T + 3TC
(n=33)

Duration of Virologic Response Definition

Duration of virologic response was defined as the time from study initiation to the time of loss of virologic response (two consecutive HIV RNA measurements above 400 copies/mL following any measurement below 400 copies/mL). If the final measurement for a patient was above 400 copies/mL (and the patient had not previously demonstrated a loss of response), the time of loss of response was defined to be the time of the last measurement. Subjects who had never experienced a loss of response were considered censored at the time of their final measurement, including subjects who prematurely discontinued prior to demonstrating a loss of response. Subjects who never achieved an HIV RNA level below 400 copies/mL were considered to have had a loss of response at Day 1.

RESULTS

Baseline Characteristics

- Ninety-six male and 4 female patients: 65% Caucasian, 29% Black, 6% Hispanic.
- Mean age: 35 years (range 21-59).
- Median Plasma HIV RNA: 4.8 $\log_{10}$ copies/mL (range 3.3-6.3).
- Median CD4 count: 326 cells/mm³ (range 3-918).

Viral Load Suppression to <400 Copies/mL at 108 Weeks

- On-treatment analysis: 99% of patients had viral load (VL) <400 copies/mL (Figure 3).
- Intent-to-treat analysis (ITT M=F; missing values considered as treatment failure): 80% of patients had VL <400 copies/mL (Figure 3).

Figure 3. M97-720 HIV RNA <400 Copies/mL Through Week 108

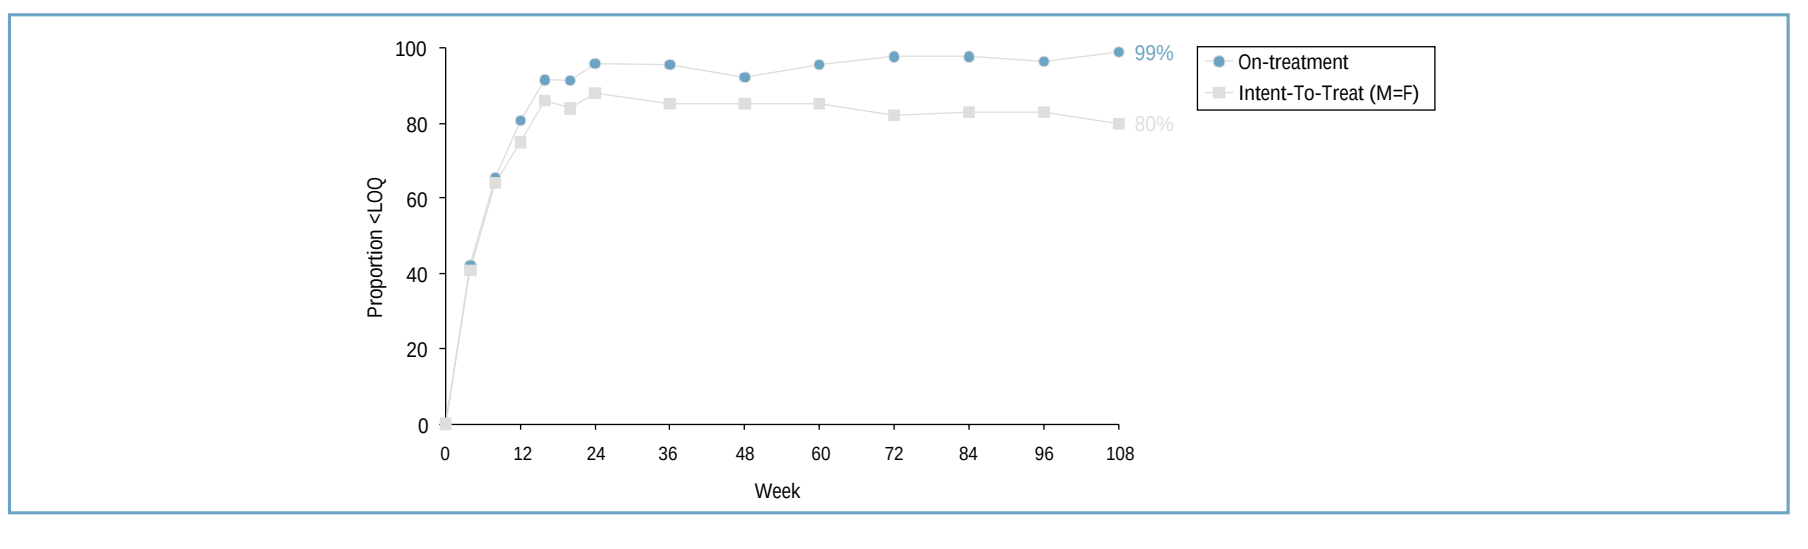
Proportion <LOQ
100
80
60
40
20
0
0
12
24
36
48
60
72
84
96
108
Week
99%
80%
On-treatment
Intent-To-Treat (M=F)

Duration of Virologic Response Analysis

- 54% (7/13) of antiretroviral-naïve patients who met loss of response criteria subsequently had viral load <400 copies/mL at the Week 108 evaluation or last visit (Figure 5).

Figure 4. M97-720: Kaplan-Meier Estimates of the Duration of Virologic Response

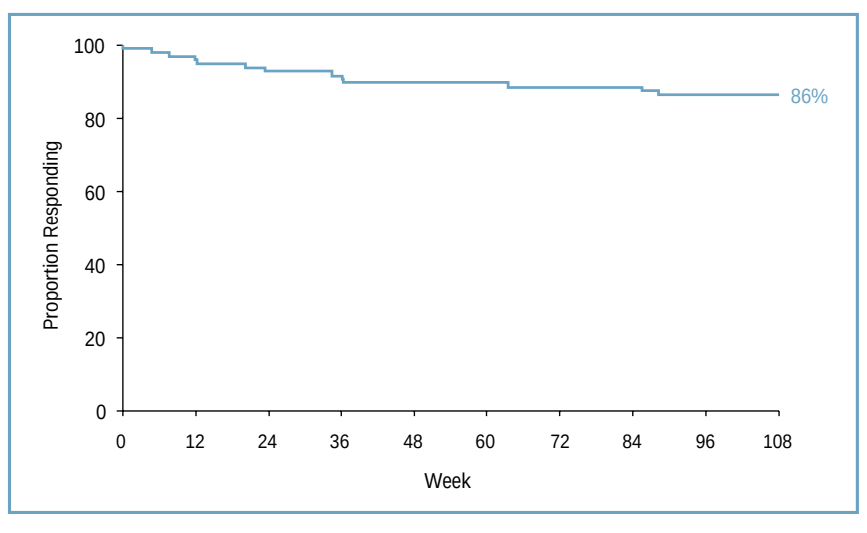
Proportion Responding
100
80
60
40
20
0
0
12
24
36
48
60
72
84
96
108
Week
86%

Figure 5. Disposition of Patients with Virologic Failure Through Week 108

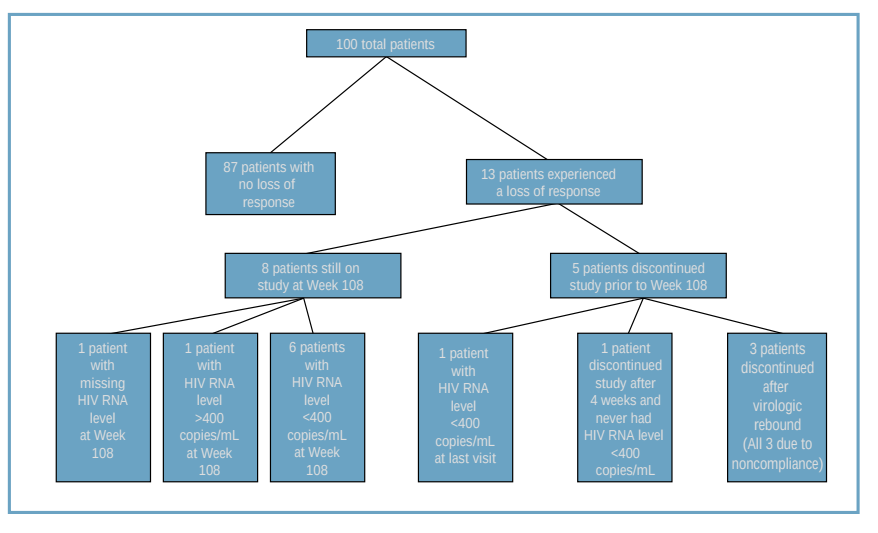
100 total patients
87 patients with no loss of response
13 patients experienced a loss of response
8 patients still on study at Week 108
5 patients discontinued study prior to Week 108
1 patient with missing HIV RNA level at Week 108
1 patient with HIV RNA level >400 copies/mL at Week 108
6 patients with HIV RNA level <400 copies/mL at Week 108
1 patient with HIV RNA level <400 copies/mL at last visit
1 patient discontinued study after 4 weeks and never had HIV RNA level <400 copies/mL
3 patients discontinued after virologic rebound (All 3 due to noncompliance)

Viral Load Suppression with Ultrasensitive Assays

- At 96 Weeks by Roche Ultrasensitive testing, HIV RNA was <50 copies/mL in 92% of patients on treatment (ITT M=F: 78%) (Figure 6).
- 56% (54/96) of patients on treatment demonstrated a viral load ≤3 copies/mL for at least one visit (Table 1).
- 72% (38/53) of patients with BL HIV RNA <100,000 copies/mL achieved a viral load ≤3 copies/mL for at least one visit compared to only 37% (16/43) of those with BL HIV RNA >100,000 copies/mL (Table 1).
- 67% (41/61) of patients with BL CD4 cell count >200 cells/µL achieved a viral load ≤3 copies/mL for at least one visit compared to only 37% (13/35) of those with BL CD4 cell counts <200 cells/µL (Table 1).

Figure 6. M97-720: HIV RNA <50 Copies/mL Through Week 96

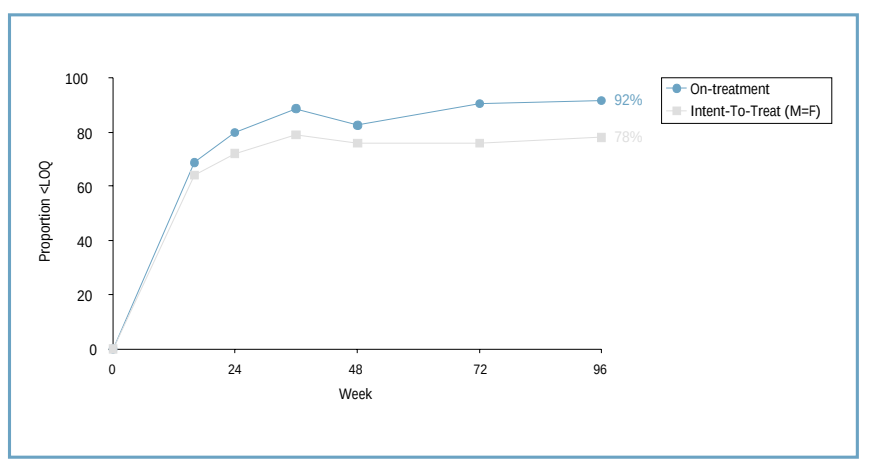
Proportion <LOQ
100
80
60
40
20
0
0
24
48
72
96
Week
92%
78%
On-treatment
Intent-To-Treat (M=F)

Table 1. M97-720: HIV RNA ≤3 Copies/mL

Type	Any Value at Weeks 24, 48, 72 ≤3 Copies/mL
Overall	54/96 (56%)
Baseline Viral Load <100,000 copies/mL	38/53 (72%)
Baseline Viral Load >100,000 copies/mL	16/43 (37%)
Baseline CD4 Cell Count >200 cells/µL	41/61 (67%)
Baseline CD4 Cell Count ≤200 cells/µL	13/35 (37%)

CD4 Cell Response

- The mean CD4 cell count at Week 108 was 599 cells/mm³ and the mean increase from baseline was 299 cells/mm³.
- The mean increase in CD4 count was consistent regardless of baseline CD4 cell count. Among 17 patients with baseline CD4 cell count <50 cells/mm³, the mean increase to Week 108 was 295 cells/mm³.

Figure 7. CD4 Cell Count (Mean Change from Baseline)

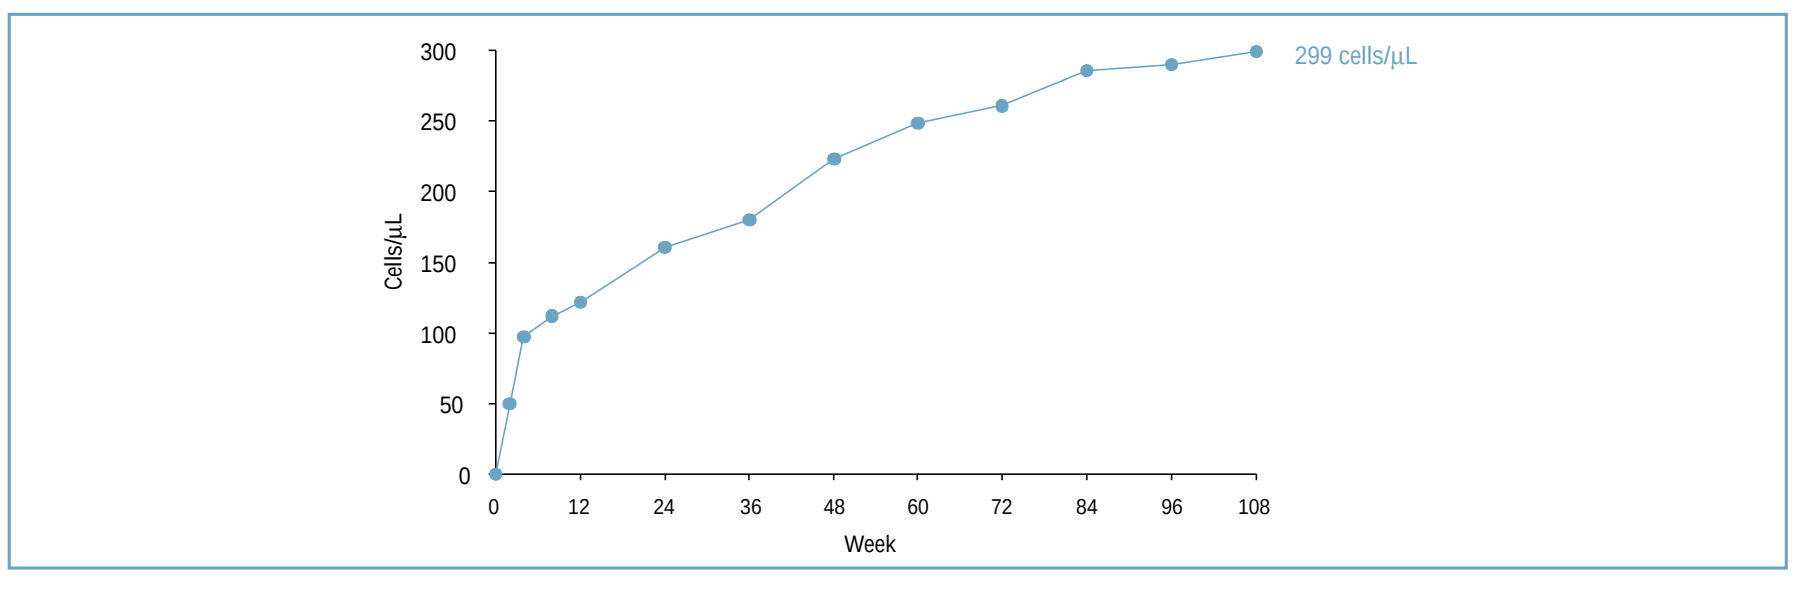
Cells/µL
300
250
200
150
100
50
0
0
12
24
36
48
60
72
84
96
108
Week
299 cells/µL

Table 2. 108-Week Safety Summary

Most Common Adverse Events* and Grade 3/4 Laboratory Abnormalities	All Patients (n=100)
Diarrhea+	24%
Nausea	15%
Abdominal Pain	8%
Abnormal Stools++	8%
Asthenia	8%
Headache	7%
Vomiting	5%
Rash	4%
Cholesterol (>300 mg/dL)	15%
Triglycerides (>750 mg/dL)	13%
AST/ALT (>5X ULN)	10%
Amylase (>2X ULN)	4%

* Adverse events of at least moderate severity and probable, possible, or unknown relationship to ABT-378/r are included.
\+ >3 loose stools/day.
++ ≤3 loose stools/day.

Table 3. Patient Disposition at Week 108

Patients enrolled	**100**
Patients discontinuing at or before Week 108	**15**
Discontinuations related to study drugs	
AST/ALT increases	1
Diarrhea	1
Arthralgia	1
Other reasons for discontinuation	
Adverse event/HIV-related event (lymphoma, hyperglycemia in diabetic patient, alcohol detoxification)	3
Personal reasons (left USA)	1
Noncompliance	4
Lost to follow-up	3
Other (drug addiction)	1

CONCLUSIONS

- ABT-378/r exhibits a potent and durable antiviral effect through 108 Weeks in antiretroviral-naïve patients, with VL <400 copies/mL in 99% of patients on treatment (ITT M=F: 80%).
- Of patients who met criteria for loss of virologic response, 54% (7/13) subsequently had a viral load <400 copies/mL at Week 108 or final evaluation.
- While over 50% of patients on ABT-378/r therapy through 72 weeks had at least one HIV RNA value ≤3 copies/mL, achieving viral suppression to this level was more likely in patients with higher CD4 counts and lower viral loads at baseline.
- ABT-378/r was well tolerated with only three patients out of 100 discontinuing due to adverse events related to study drug through 108 Weeks.
- Early use of ABT-378/r is supported by the tolerability profile and durable antiviral activity observed to date.

ACKNOWLEDGMENTS

M97-720 Study Subjects	
Covance Central Laboratory Services	
AIDS Research Consortium of Atlanta	*Sullivan M, Bohn H, Enstrom T*
Beth Israel Deaconess Medical Center	*Fitch H, Koziol C*
Cornell Clinical Trials Unit	*Sarraco T, Stroberg T*
Duke University Medical Center	*Giner J, Harmon L*
Northwestern University	*Werry A, Bruce J, Donath P*
Pacific Oaks Research	*Perry B, Walker S*
Rush Presbyterian St. Luke's Medical Center	*Narkiewicz E*
Thomas Street Clinic	*Sepcie B*
University of Colorado	*Canmann S, Putnam B, Ray MG*
University of North Carolina at Chapel Hill	*Marcus C, Ngo L*
PPD Development	*McCarley S, Donnelly A, Jackson S, Nicks B*
Abbott Laboratories	*Kempf D, Potthoff A, Rode R, Sheehan K, Yang G*
Laboratory of Virology, U. Geneva	*Yerly S, Wegmann L*

REFERENCES

1. Bertz R, Lam W, Brun S, et al. Multiple-dose pharmacokinetics (PK) of ABT-378/ritonavir (ABT-378/r) in HIV+ subjects. 39th Interscience Conference on Antimicrobial Agents and Chemotherapy, San Francisco, USA, 1999 (abstract 0327).
2. Kempf DJ, Molla A, Hsu A. Protease inhibitors as anti-HIV agents. Antiretroviral Therapy, American Society for Microbiology Press, E DeClerq, editor. In press.
3. Ritonavir Package Insert.
4. Indinavir Package Insert.
5. Amprenavir Package insert.
6. Nelfinavir Package Insert.
7. Saquinavir (Fortavase) Package Insert.
8. Cohen C, et al. Potent and convenient Fortovase™ (SQV) SGC BID regimens in combination with 2 nucleosides or nelfinavir (NFV) plus 1 nucleoside in HIV-1 infected patients. 12th World AIDS Conference, Geneva, Switzerland, 1998, (abstract 12314).
9. Saag M, et al. Saquinavir systemic exposure and safety on once daily administration of Fortovase™ (Saquinavir) soft gel capsules (FTV) in combination with low-dose ritonavir. 39th Interscience Conference on Antimicrobial Agents and Chemotherapy, San Francisco, USA, 1999, (abstract 330).
10. Shulman N, et al. Ritonavir intensification in indinavir recipients with detectable HIV RNA levels. 7th Conference on Retroviruses and Opportunistic Infections, San Francisco, USA (abstract 534).
11. Piscitelli S, et al. The addition of a second protease inhibitor eliminates Amprenavir-Efavirenz drug interactions and increases plasma Amprenavir concentrations. 7th Conference on Retroviruses and Opportunistic Infections, San Francisco, USA (abstract 78).
12. Mallolas J, et al. A dose-finding study of once-daily Indinavir/Ritonavir plus Combivir® in HIV infected patients. First International Workshop on Clinical Pharmacology of HIV Therapy, March 2000 (abstract 2.14).
13. Flexner C, et al. Steady-state pharmacokinetic interactions between ritonavir (RTV), nelfinavir (NFV), and the nelfinavir active metabolite M8 (AG1402). 12th World Aids Conference, Geneva (abstract 42265).
14. Cameron DW, et al. Ritonavir and saquinavir combination therapy for the treatment of HIV infection. AIDS, 1999, 12(2) 213-224.
15. Yerly S., Rutschmann O.T., Perrin L. et al. Cell-Associated HIV-1 RNA in Blood as Indicator of Virus Load in Lymph Nodes. The Journal of Infectious Diseases, 1999;180:50-3.

*Amplicor HIV-1 Monitor is a trademark of Roche Molecular Diagnostics

Kaletra™ is not yet licensed in the U.K.

E002404F